Tethered IBD Tools to Support Patients and Providers in Shared Decision-Making and Therapy Access

Raymond Cross[1], Ellen Scherl[2], Kristina Fajardo[3], Eric Weinbaum[3], Laurence Greene[3], Paula Eichenbrenner[4], Kathleen Fraser[5], Ali Shipley Bender[6], Kathleen Moreo[3]

(1) University of Maryland School of Medicine, Baltimore, MD, (2) Weill Cornell Medical College of Cornell University, New York, NY (3) PRIME Education, LLC, Fort Lauderdale, FL, (4) AMCP Foundation, Alexandria, VA, (5) Case Management Society of America, Little Rock, AR, (6) CCFA Support Group Facilitator and Education Events Committee Member, Coral Springs, FL.

#P011

INTRODUCTION

Advances in inflammatory bowel disease (IBD) treatment have improved outcomes for many patients[1]. However, gaps at the patient, provider, and system level often limit the extent to which patients benefit from these advances. These barriers include, but are not limited to:

- Suboptimal patient and provider knowledge about evidence-based IBD treatment[2]
- Inaccurate, misleading, or outdated medical information accessed by patients online[3]
- Discordance between patient and provider attitudes and perceptions on IBD care and treatment[4]
- Challenges in navigating managed care processes for therapy access

Effective tools are needed to support IBD patients and providers in shared decision-making (SDM) and navigating access to appropriate therapies[5,6]. We developed and evaluated patient and provider educational online apps that present aligned content designed to close gaps in SDM and timely treatment.

METHODS

- We developed patient and provider web apps that present aligned content on evidence-based IBD treatment recommendations, combined with tools and strategies, for applying SDM and navigating managed care processes.
- The Web Apps were reviewed by leading IBD experts, an IBD patient advocate, and representatives from the Academy of Managed Care Pharmacy Foundation and the Case Management Society of America to ensure relevancy across stakeholders.
- Pre- and post-education survey items were embedded to evaluate the effectiveness of the tools and assess patient and provider perceptions of IBD treatment and management.
- Users navigating through the web app can bookmark relevant information, tips, and resources to a personalized handout for download.
- Browser-based experience (vs. native smartphone app) ensures easy access on any Internet-enabled device without needing to download any software or register an account.

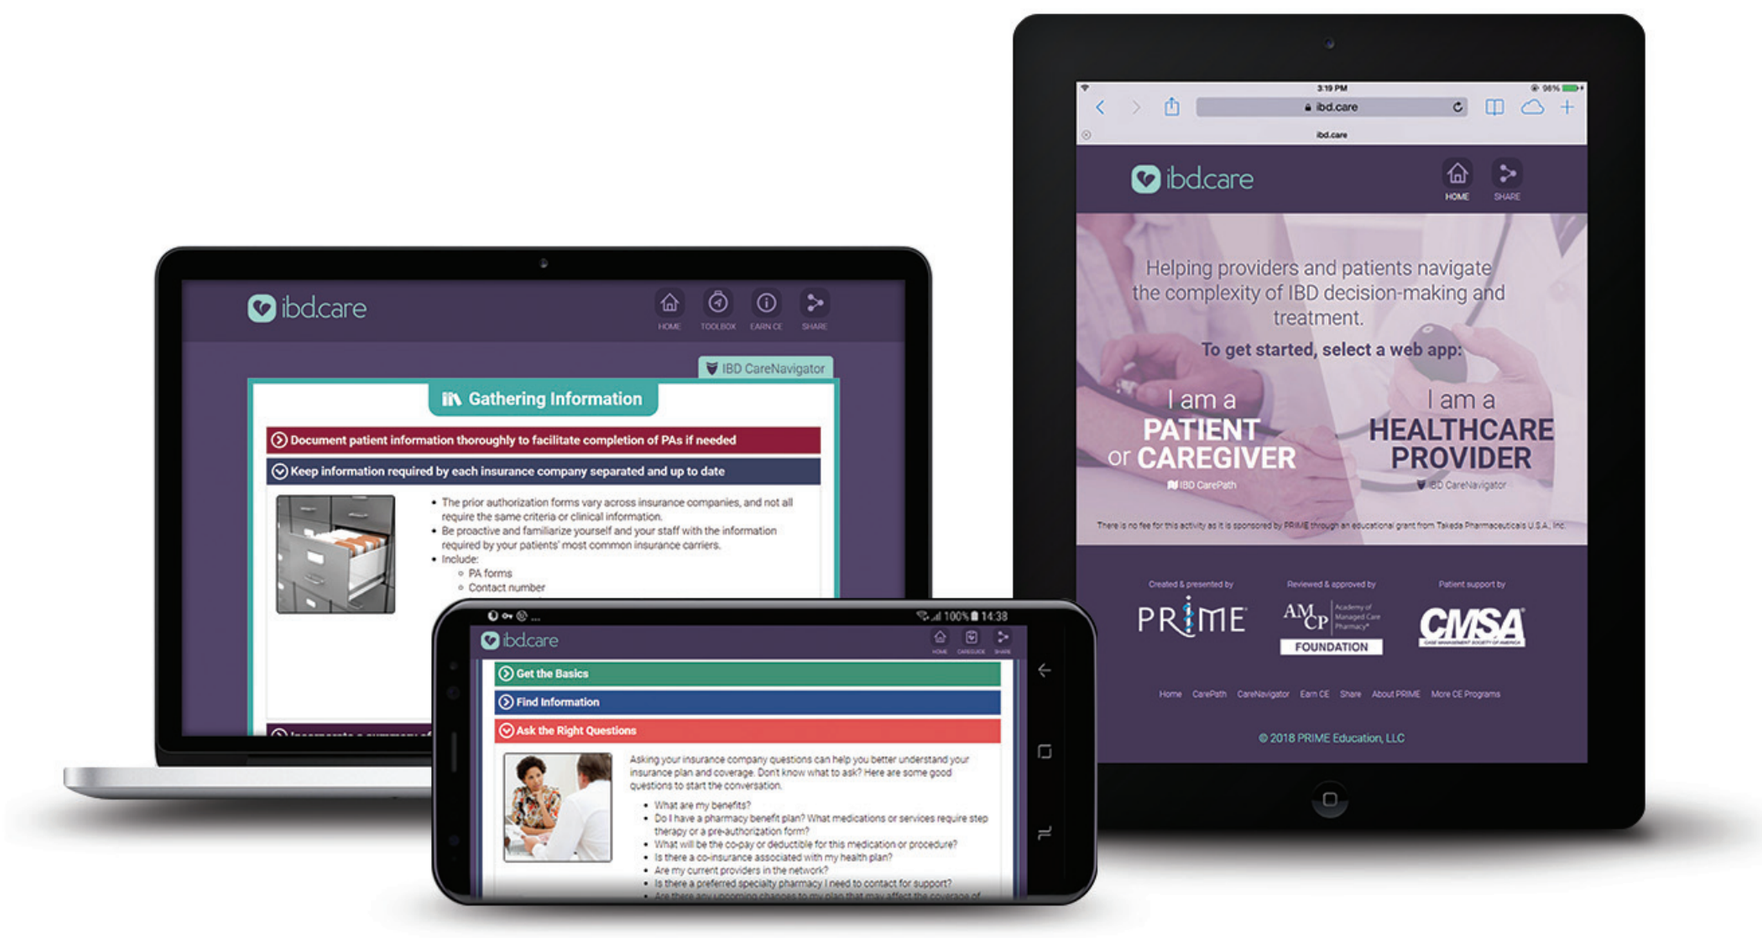

ibdcare
Gathering Information
Helping providers and patients navigate the complexity of IBD decision-making and treatment.
To get started, select a web app:
I am a PATIENT or CAREGIVER
I am a HEALTHCARE PROVIDER
Get the Basics
Find Information
Ask the Right Questions

The web apps were launched in February 2018. Results reflect data collected from 2/14/2018 to 1/15/2019. Collection of data is ongoing.

Access now at **www.ibd.care**

RESULTS

TABLE 1: WEB APP USER METRICS

	Patient	Provider
Unique Users	7,328	5,647
Returning Users	2,428	1,251
Survey Respondents	384 – 561	189 – 592
Handout Downloads	364	

TABLE 2: DEMOGRAPHICS

Patients (N = 353)	
% Female	68%
Mean Age (Years)	44
Crohn's Disease	38%
Ulcerative Colitis	41%
Do not know	21%
IBD Disease Duration (years)	9

Providers (N = 1,064)	
Physician	28%
Nurse Practitioner or Physician Assistant	15%
Nurse/Nurse Navigator	52%
Medical Assistant	5%

PATIENT SELF-REPORTED PERCEPTIONS (N = 353)

44% Experience high or very high disruption of day-to-day activities due to IBD symptoms

38% Have experienced insurance denial of IBD medication(s) prescribed by provider

35% Have high or very high understanding of their insurance coverage

37% Receive high / very high support from clinicians in navigating insurance coverage

28% Experience >2 week delays from prescription to insurance coverage approval of therapy

PROVIDER SELF-REPORTED MEASURES (N = 1,064)

Ratings of 4 or 5 on a 5-point Likert scale

PRE-ACTIVITY / POST-ACTIVITY

76% → 84% — **High/Very High Agreement**: Using evidence-based IBD care pathways within SDM supports informed treatment decisions

17% → 36% — **High/Very High Frequency**: Currently use /intend to use AGA Care Pathways to guide IBD treatment decisions

21% → 46% — **High/Very High Confidence**: Navigating prior authorization requirements for IBD therapies and procedures

PATIENT AND PROVIDER ESTIMATES ON PATIENT'S: PREFERENCE FOR BEING "HIGHLY INVOLVED" IN SDM

PROVIDERS (N = 1,064): 40%

PATIENTS (N = 353): 66%

TOP TREATMENT GOALS

PROVIDERS
- Achieve symptom-free remission
- Improve quality of life
- Avoid hospitalizations and surgery

PATIENTS
- Maintain symptom-free remission
- Avoid frequent use of steroids
- Avoid hospitalizations and surgery

PROVIDER POST-PROGRAM COMMITMENT TO IMPROVE CARE (N = 1,064)

47% IMPROVE DOCUMENTATION OF PATIENT INFORMATION

38% KEEP COPIES OF PATIENTS' INSURANCE REQUIREMENTS

35% SUBMIT PRIOR AUTHORIZATIONS ELECTRONICALLY

29% ENROLL OR TRAIN STAFF TO OVERSEE PA PROCESS

CONCLUSIONS

Key findings from the baseline patient and provider survey items include:

- Providers underestimated the proportion of patients who would like to be highly involved in IBD treatment decisions
- While providers recognized that using evidence-based pathways is important for making informed decisions, a minority used or planned to use the AGA Care Pathways
- Patients and providers had limited confidence in and understanding of managed care processes
- Patients reported experiencing barriers to accessing needed IBD therapies, including denials of prescribed therapy and delays in therapy approval

The web apps present a model for patient and provider education aligned with the principles of SDM, and may be useful to encourage adoption of treatment guidelines in PAs, electronic PAs, and EHR management.

REFERENCES

1. Duijvestein M, et al. *Curr Treat Options Gastroenterol.* 2018;16(1):129–146.
2. Sapir T, et al. *Dig Dis Sci.* 2016 Jul;61(7):1862–1869
3. Martinez B, et al. *Inflamm Bowel Dis.* 2017;23(7):1057–1064.
4. Schreiber S, et al. *BMC Gastroenterol.* 2012;12:108.
5. Almario CV, et al. *Am J Gastroenterol.* 2018;113(1): 58.
6. American Medical Association. *Digital Health Study.* 2016. Accessed at: www.ama-assn.org/sites/default/files/media-browser/specialty%20group/washington/ama-digital-health-report923.pdf

DISCLOSURES

Raymond K **Cross**, MD, MS, AGAF, FACG is an advisory board or panel member, consultant, and principle investigator on a research grant for AbbVie; as well as is an advisory board or panel member for Janssen and Pfizer. Ellen J **Scherl**, MD, FACP, FACG, AGAF, FASGE, NYSGEF is an advisory board or panel member and consultant for AbbVie, Entera Health, Evidera, GI Health Foundation, Janssen, Protagonist Therapeutics, Seres Health, and Takeda Pharmaceuticals; as well as principle investigator on a research grant for Abbott, AstraZeneca, CCFA, Janssen Research & Development, Millennium Pharmaceuticals, National Institute of Diabetes and Digestive and Kidney (NIDDK), National Institute of Health (NIH), Pfizer, Prometheus Laboratories, Salix, UCB, and UCSF-CCFA Clinical Research Alliance; and a self-managed stock/shareholder of Gilead. There are no relationships to disclose for Kristina **Fajardo**, MS; Eric **Weinbaum**, BS; Laurence **Greene**, PhD; Paula **Eichenbrenner**; Kathleen **Fraser**; Ali **Shipley Bender**; and Kathleen **Moreo**, RN-BC, BSN, BHSA, CCM, CDMS.

This project was funded by an independent educational grant from Takeda Pharmaceuticals U.S.A. The study sponsor did not play a role in the design or analysis of the study or in the decision to submit for presentation.

Contact: l.greene@primeinc.org